Incorporating Analytical Variance Into a Comparative Quantitative Risk Assessment (QRA) Approach for Tobacco Products

Chastain A. Anderson, Vanessa Haase, Kimberly D. Ehman, Paige N. Wiecinski, Donna C. Smith
Altria Client Services LLC, Richmond, VA 23219

73rd TSRC - Tobacco Science Research Conference
September 15-18, 2019, Leesburg, VA, USA

WWW.ALTRIA.COM/ALCS-SCIENCE | Altria

ABSTRACT

The US Food and Drug Administration (FDA) Draft Guidance for Industry (2012) provides abbreviated lists of harmful and potentially harmful constituents (HPHCs) found in tobacco and cigarette smoke. As FDA considers the abbreviated HPHC list to be representative of the classes of hazardous compounds present in tobacco products or tobacco smoke, it can serve as the basis of a relative "whole-product" toxicological risk comparison for tobacco products (e.g., New Product to Predicate Product). A range of validated analytical techniques exist for quantification of individual HPHCs, which are reported as a mean value with a standard deviation. The objective of this presentation is to propose a method for incorporating analytical variance into a comparative quantitative risk assessment (QRA) framework to provide a more robust representation of relative toxicological risk between whole tobacco products. Using a cigarette example, we provide a general overview of the QRA framework (i.e., hazard assessment, exposure assessment, and risk characterization) and highlight how incorporating the analytical variance in HPHC yields into the exposure assessment and hazard characterization produces an estimate of the range of probable risk as opposed to a single (deterministic) point estimate. The range of risk for each individual HPHC is calculated for both cancer (i.e., Excess Lifetime Cancer Risk – ELCR) and non-cancer (i.e., Hazard Index – HI) effects, and these individual risk estimates are then aggregated into representative estimates of whole-product risk. Additionally, statistical analysis can be performed to assess whether there is a significant difference ($p < 0.05$) in risk between the products.

METHODOLGY

- Marano, et al. 2018, demonstrated that US Environmental Protection Agency risk assessment guidelines (US EPA, 1989) can be used to compare tobacco products on the basis of point estimates of risk derived from HPHCs. Here, we expand this approach to incorporate the analytical variance in HPHC measurements and a statistical analysis of whole-product risk.
- This approach compares the relative, representative risk of a New Product and a Predicate Product to determine whether they significantly differ. The relative risk is used for comparative purposes only and should not be mistaken for the absolute risk of any single product.
- The risk assessment of each individual HPHC was performed in a five-step process:
 1. Hazard Assessment
 2. Exposure Assessment
 3. Individual Risk Characterization
 4. Representative Whole-Product Risk Characterization
 5. Statistical Analysis

HAZARD ASSESSMENT

- We conducted a comprehensive review of regulatory values for each member of the abbreviated HPHC list to source regulatory values for both cancer and non-cancer risk.

 The priority tier for sourcing regulatory values was:
 1. US EPA Integrated Risk information System (IRIS)
 2. California Environmental Protection Agency (Cal EPA)
 3. Texas Commission on Environmental Quality (TCEQ)
 4. WHO Guidelines for Indoor Air Quality
- The majority of the values included in this assessment are US EPA IRIS inhalation unit risks (IUR) or reference concentrations (RfC).

EXPOSURE ASSESSMENT

- An estimated exposure concentration was generated per (US EPA) guidance on estimating chemical intakes (US EPA 1989).
- Conventional estimates of chemical intake generate a single point estimate. However, machine measured HPHC yield is normally distributed and approximated by the sample mean ($\bar{x}$) and sample standard deviation (s).
- For each individual HPHC, the yield per cigarette ($\bar{x}$,s) was multiplied by an estimate of consumption rate in cigarettes per day, an estimate of years spent smoking, and the number of days per year. This value was averaged over an estimate of total inhaled air for the duration of exposure to generate a final estimated exposure concentration (EC) in μg/m³.

$$EC = \frac{C_{(\bar{x},s)} \times CpD \times ED \times EF}{IR \times AT}$$

EXPOSURE ASSESSMENT CONT'D

Parameter	Symbol	Value	Unit	Source
HPHC Measured Yield	$C_{(x,s)}$	variable	μg/cigarette	Internal
Cigarette Consumption	*CpD*	14.1	cigarettes/day	CDC (2018)
Exposure Duration	*ED*	57.5	years	US FDA (2013)
Exposure Frequency	*EF*	365	days/year	Maximum value
Inhalation Rate	*IR*	20	m³/day	US EPA (1991, 2011)
Average Time	*AT*	20,988	days	US EPA (1989, 2009, 2014)

HPHC	New Product		Predicate Product	
	Yield (μg/cigarette)	Exposure (μg/m3)	Yield (μg/cigarette)	Exposure (μg/m3)
Acetaldehyde	523.4 ± 54.2	369 ± 38.2	512.8 ± 49	361.5 ± 34.5
Acrolein	49.2 ± 4.55	34.7 ± 3.21	47.9 ± 4.4	33.8 ± 3.1
Acrylonitrile	7.66 ± 0.485	5.4 ± 0.342	7.53 ± 0.487	5.31 ± 0.343
Ammonia	9.31 ± 0.553	6.56 ± 0.39	9.04 ± 0.507	6.37 ± 0.357
Benzene	34.7 ± 2.21	24.5 ± 1.56	33.8 ± 1.94	23.8 ± 1.37
Benzo[a]pyrene	0.0049 ± 0.000138	0.00345 ± 0.0000973	0.00484 ± 0.00013	0.00341 ± 0.0000916
1,3-Butadiene	34.4 ± 1.687	24.3 ± 1.19	33.6 ± 1.72	23.7 ± 1.21
Carbon Monoxide	9951.5 ± 710.5	7015.6 ± 500.9	9712.6 ± 664.4	6847.2 ± 468.4
Crotonaldehyde	10.9 ± 1.62	7.68 ± 1.14	10.6 ± 1.46	7.47 ± 1.03
Formaldehyde	27.4 ± 2.6	19.3 ± 1.83	26.6 ± 2.52	18.8 ± 1.78
Isoprene	317.2 ± 15.5	223.6 ± 10.9	306.2 ± 16	215.9 ± 11.3
NNK	0.0959 ± 0.00818	0.0676 ± 0.00577	0.0932 ± 0.00812	0.0657 ± 0.00572
NNN	0.0925± 0.00243	0.0652 ± 0.00171	0.0895 ± 0.0027	0.0631 ± 0.0019
Toluene	53.8 ± 3.63	37.9 ± 2.56	51.8 ± 3.38	36.5 ± 2.38

Note – HPHC yields presented here have been abstracted to be within the range of yields from actual products but are not representative of any single product or smoking regimen.

HPHC	IUR (μg/m³)⁻¹	ELCR		RfC (mg/m³)	HQ	
		New	Predicate		New	Predicate
Acetaldehyde	2.20E-06[1]	8.12E-04 ± 8.40E-05	7.95E-04 ± 7.59E-05	9.00E-03[1]	4.10E+01 ±4.24E+00	4.02E+01 ± 3.83E+00
Acrolein	NA	NA	NA	2.00E-05[1]	1.74E+03 ±1.61E+02	1.69E+03 ± 1.55E+02
Acrylonitrile	6.80E-05[1]	3.67E-04 ± 2.33E-05	3.61E-04 ± 2.33E-05	2.00E-03[1]	2.70E+00 ± 1.71E-01	2.66E+00 ± 1.72E-01
Ammonia	NA	NA	NA	5.00E-01[1]	1.31E-02 ± 7.80E-04	1.27E-02 ± 7.14E-04
Benzene	7.80E-06[1]	1.91E-04 ± 1.22E-05	1.86E-04 ± 1.07E-05	3.00E-02[1]	8.17E-01 ± 5.20E-02	7.93E-01 ± 4.57E-02
Benzo[a]pyrene	6.00E-04[1]	2.07E-06 ± 5.84E-08	2.05E-06 ± 5.50E-08	2.00E-06[1]	1.73E+00 ± 4.87E-02	1.71E+00 ± 4.58E-02
1,3-Butadiene	3.00E-05[1]	7.29E-04 ± 3.57E-05	7.11E-04 ± 3.63E-05	2.00E-03[1]	1.22E+01 ± 5.95E-01	1.19E+01 ± 6.05E-01
Carbon Monoxide	NA	NA	NA	7.00E+00[4]	1.00E+00 ± 7.16E-02	9.78E-01 ± 6.69E-02
Crotonaldehyde	NA	NA	NA	4.00E-03[3]	1.92E+00 ± 2.85E-01	1.87E+00 ± 2.58E-01
Formaldehyde	1.30E-05[1]	2.51E-04 ± 2.38E-05	2.44E-04 ± 2.31E-05	9.00E-03[3]	2.14E+00 ± 2.03E-01	2.09E+00 ± 1.98E-01
Isoprene	2.20E-08[2]	4.92E-06 ± 2.40E-07	4.75E-06 ± 2.49E-07	3.90E-01[2]	5.73E-01 ± 2.79E-02	5.54E-01 ± 2.90E-02
NNK	1.40E-02[3]	9.46E-04 ± 8.08E-05	9.20E-04 ± 8.01E-05	NA	NA	NA
NNN	4.00E-04[3]	2.61E-05 ± 6.84E-07	2.52E-05 ± 7.60E-07	NA	NA	NA
Toluene	NA	NA	NA	5.00E+00[1]	7.58E-03 ± 5.12E-04	7.30E-03 ± 4.76E-04

[1]US EPA IRIS, [2]Cal EPA, [3]TCEQ, [4]WHO, NA – Not Applicable

RISK CHARACTERIZATION

- Risk characterization combines the values from the hazard assessment and the exposure assessment to produce a risk estimate. The two types of risk estimates used in this assessment are excess lifetime cancer risk (ELCR) and hazard quotient (HQ).
 - ELCR represents the incremental probability of an individual developing cancer over a lifetime under the specified exposure conditions. For example, an ELCR of 0.0001 would represent a life time probability of 1 excess cancer case per 10,000 individuals.
 - The HQ represents adverse health events (e.g., local or systemic effects) with an established threshold (e.g. US EPA RfC). An HQ less than 1 indicates that the estimated exposure concentration is less than the established safety threshold (i.e. is below a level of toxicological concern). If the HQ is above 1, then exposure to the HPHC is introducing some level of non-cancer risk.

ELCR is calculated by the following equation:

$ECLR_{(\bar{x},s)} = EC_{(\bar{x},s)} \times IUR$, where:

- ELCR– estimated lifetime cancer risk (unitless)
- EC – exposure concentration (μg/m³)
- IUR – Inhalation Unit Risk $(\mu g/m^3)^{-1}$

HQ is calculated by the following equation:

$HQ_{(\bar{x},s)} = \frac{EC_{(\bar{x},s)}}{RfC}$ where:

HQ – hazard quotient (unitless)
EC – exposure concentration (μg/m³)
RfC – Reference Concentration (mg/m³)

RISK CHARACTERIZATION-WHOLE PRODUCT

- As the normally distributed risk estimates (ELCR and HQ) are weighted by an estimate of potency (IUR and RfC), the principle of dose additivity states that they can be aggregated into a cumulative ELCR and hazard index (HI) for the whole product. These whole-product risk estimates provide the basis for comparison of the relative risk of the New Product and the Predicate Product.
- Multiple normal distributions can aggregated by taking the simple sum of their means and the root sum of squares of their standard deviations.

- $cumulative\ ELCR_{\bar{x}} = \sum_{i=1}^{n} ELCR_{(\bar{x})i}$
- $HI_{\bar{x}} = \sum_{i=1}^{n} HQ_{(\bar{x})i}$
- $cumulative\ ELCR_{s} = \sqrt{\sum_{i=1}^{n} ELCR_{(s)i}^2}$
- $HI_{s} = \sqrt{\sum_{i=1}^{n} HQ_{(s)i}^2}$

INCORPORATION OF ANALYTICAL VARIANCE

- We tested the hypothesis that the representative risk of the New Product significantly differs from the representative risk of the Predicate Product. Analysis was performed in R (R Foundation for Statistical Computing, 2012) using the "tsum.test()" function from the Basic Statistics and Data Analysis (BSDA) library (Arnholt and Evans, 2017) to perform a Welch's t-test on the whole-product risk estimates.
- Despite the difference in point estimates of whole-product risk, when we incorporate the analytical variance of the machine measured yield of the HPHCs, the whole-product risk does not significantly differ ($p > 0.05$).

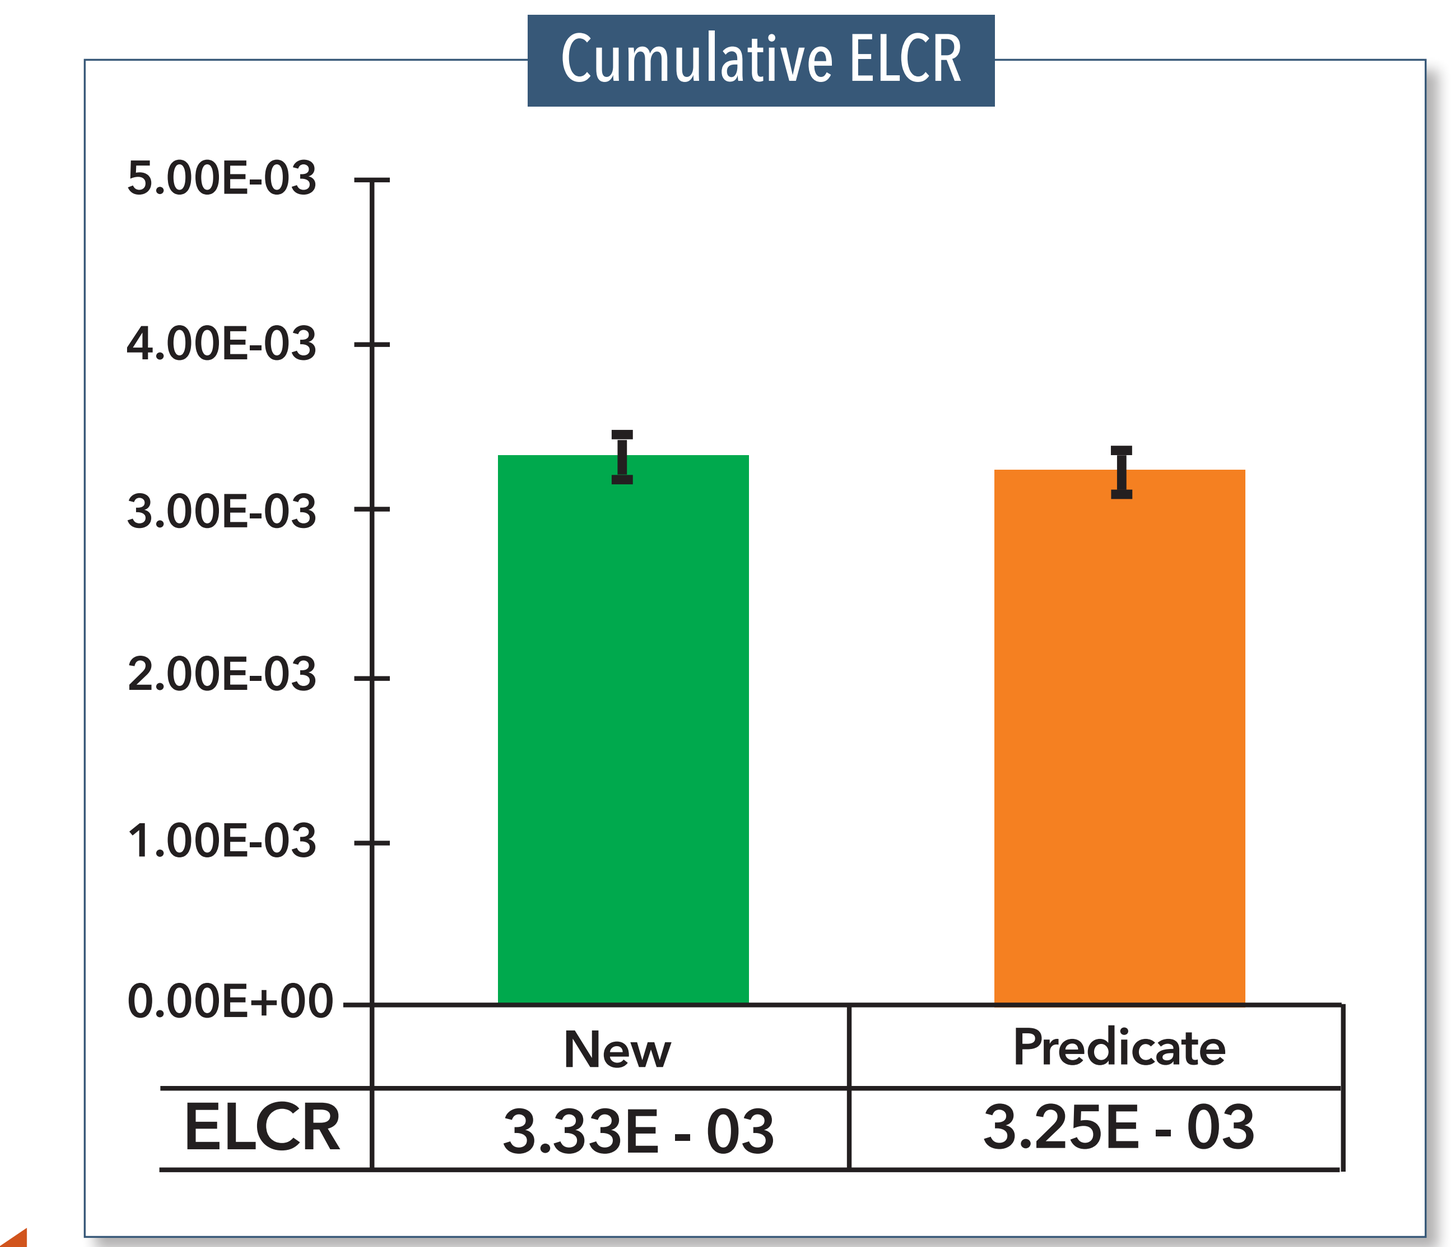
Cumulative ELCR
5.00E-03
4.00E-03
3.00E-03
2.00E-03
1.00E-03
0.00E+00
New
Predicate
ELCR
3.33E - 03
3.25E - 03

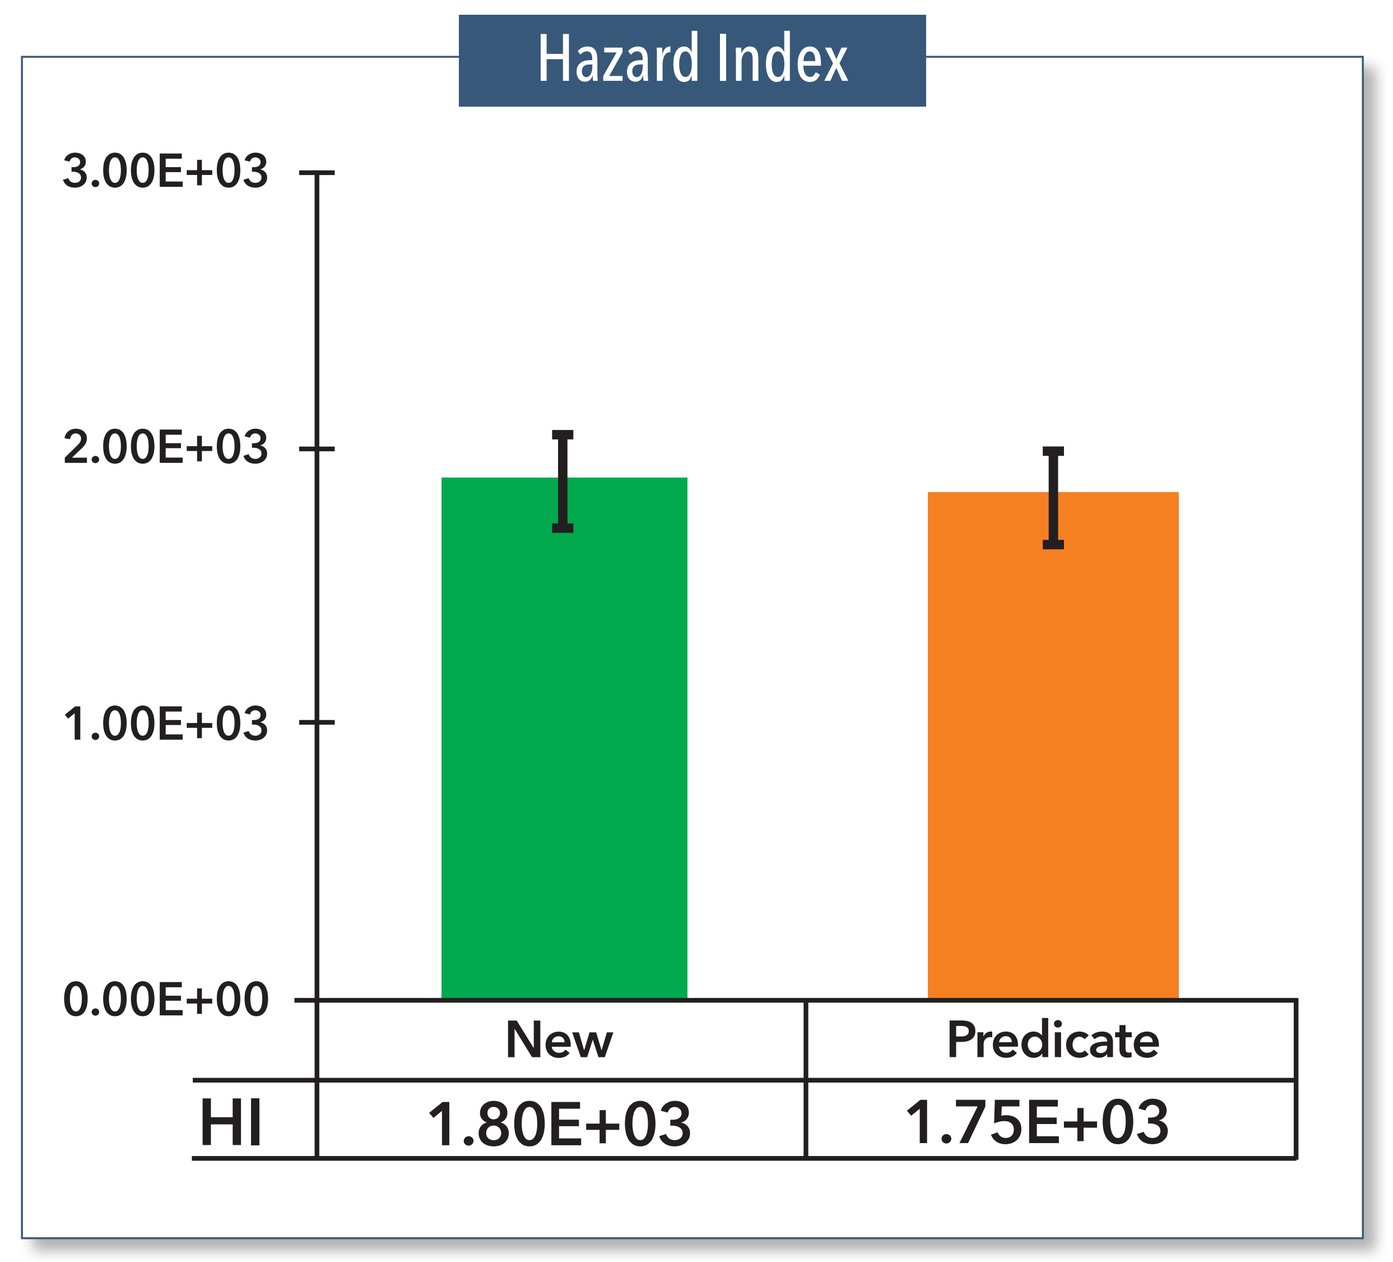
Hazard Index
3.00E+03
2.00E+03
1.00E+03
0.00E+00
New
Predicate
HI
1.80E+03
1.75E+03

STRENGTHS AND LIMITATIONS

- Strengths
 - We have adapted and expanded the Marano et al. (2018) approach to evaluate the relative risk of tobacco products.
 - While the original method calculated point estimates of risk, our approach incorporates the analytical variance in HPHC yield and statistically analyzes the difference between whole-product risk estimates.
 - While conventional tobacco cigarettes were used here as an example, this approach is broadly applicable and appropriate for any comparison between tobacco products.
- Limitations
 - A relative risk comparison is dependent on the HPHCs included in the assessment. The underlying assumption of the presented approach is that the abbreviated HPHC list for cigarettes is representative of whole-product risk.
 - Approaches are currently lacking for evaluation of HPHCs that are classified by FDA as carcinogens but lack an established IUR.
 - While this approach incorporates analytical variance in machine measured smoke yield, it still relies on point estimates of many other exposure factors.

REFERENCES

Arnholt and Evans, 2017. Basic Statistics and Data Analysis Package
Marano, K.M, Liu, C., Fuller, W, Gentry, P.R. 2018. Quantitative risk assessment of tobacco products: A potentially useful component of substantial equivalence evaluations. Regulatory Toxicology and Pharmacology 95, 371-384
R Development Core Team, 2012. R: A Language and Environment for Statistical Computing. R Foundation for Statistical Computing
US EPA, 1989. Risk Assessment Guidance for Superfund Volume 1 Human Health Evaluation Manual (Part A). Interim Final. United States Environmental Protection Agency, Office of Emergency and Remedial Response, Washington, DC EPA/540/1–89/002.
US EPA, 1991. Risk Assessment Guidance for Superfund Volume I – Human Health Evaluation Manual (Part B, Development of Risk-based Preliminary Remediation Goals) Interim. United States Environmental Protection Agency EPA/540/R-92/003. December 1991.
US EPA, 2009. Risk Assessment Guidance for Superfund Volume 1 Human Health Evaluation Manual (Part F, Supplemental Guidance for Inhalation Risk Assessment). United States Environmental Protection Agency, Office of Superfund Remediation and Technology Innovation, Washington, DC EPA-540-R-070–002.
US EPA, 2011. Exposure Factors Handbook: 2011 Edition. United States Environmental Protection Agency. Available at: https://cfpub.epa.gov/ncea/risk/recordisplay.cfm? deid=236252.
US EPA, 2014. Office of Solid Waste and Emergency Response Directive 9200.1-120. Memorandum, Human Health Evaluation Manual, Supplemental Guidance, Update of Standard Default Exposure Factors. United States Environmental Protection Agency. Available at: https://rais.ornl.gov/documents/OSWER_Directive_9200.1-120_ExposureFactors_corrected2.pdf.
US FDA, 2012. Reporting harmful and potentially harmful constituents in tobacco products and tobacco smoke under Section 904(e) of the Federal Food, Drug, and Cosmetic Act. Draft Guidance. March. United States Food and Drug Administration. Available at: http://www.fda.gov/downloads/TobaccoProducts/GuidanceComplianceRegulatoryInformation/UCM297828.pdf.
US FDA, 2013. US Food and Drug Administration, Toxicology Review of 905(j)(1)(A)(i) Report, Second Cycle Review of Additional Information, SE0003730, SE0003731. United States Food and Drug Administration Document received via Freedom of Information Act request.
CDC, 2018. Centers for Disease Control. Morbidity and Mortality Weekly Report. Current Cigarette Smoking Among Adults – United States, 2016. Vol. 67:2.